CER-001, an Engineered High-Density Lipoprotein, shows beneficial pleiotropic effects in patients with sepsis in RACERS: a Phase 2a randomized clinical trial

M. Fiorentino, A. Stasi, R. Franzin, L. Gesualdo

Nephrology Unit, Department of Precision and Regenerative Medicine and Ionian Area (DiMEPRE-J), Università degli Studi di Bari "Aldo Moro", Bari, ITALY

Financial Disclosures

This work was supported by a research grant from Abionyx Pharma to L.Gesualdo (Granted to his University Department to pay salary for A.Stasi and R.Franzin and supplies).

Background

Sepsis is characterized by a dysregulated immune response and metabolic alterations, including decreased High-Density Lipoprotein cholesterol (HDL-C) levels. Our recent research highlighted the diverse properties of HDL, including LPS scavenging, anti-inflammatory effects, and preservation of endothelial integrity in an LPS-induced AKI swine model treated with CER-001, an engineered HDL mimetic (**Figure 1**).

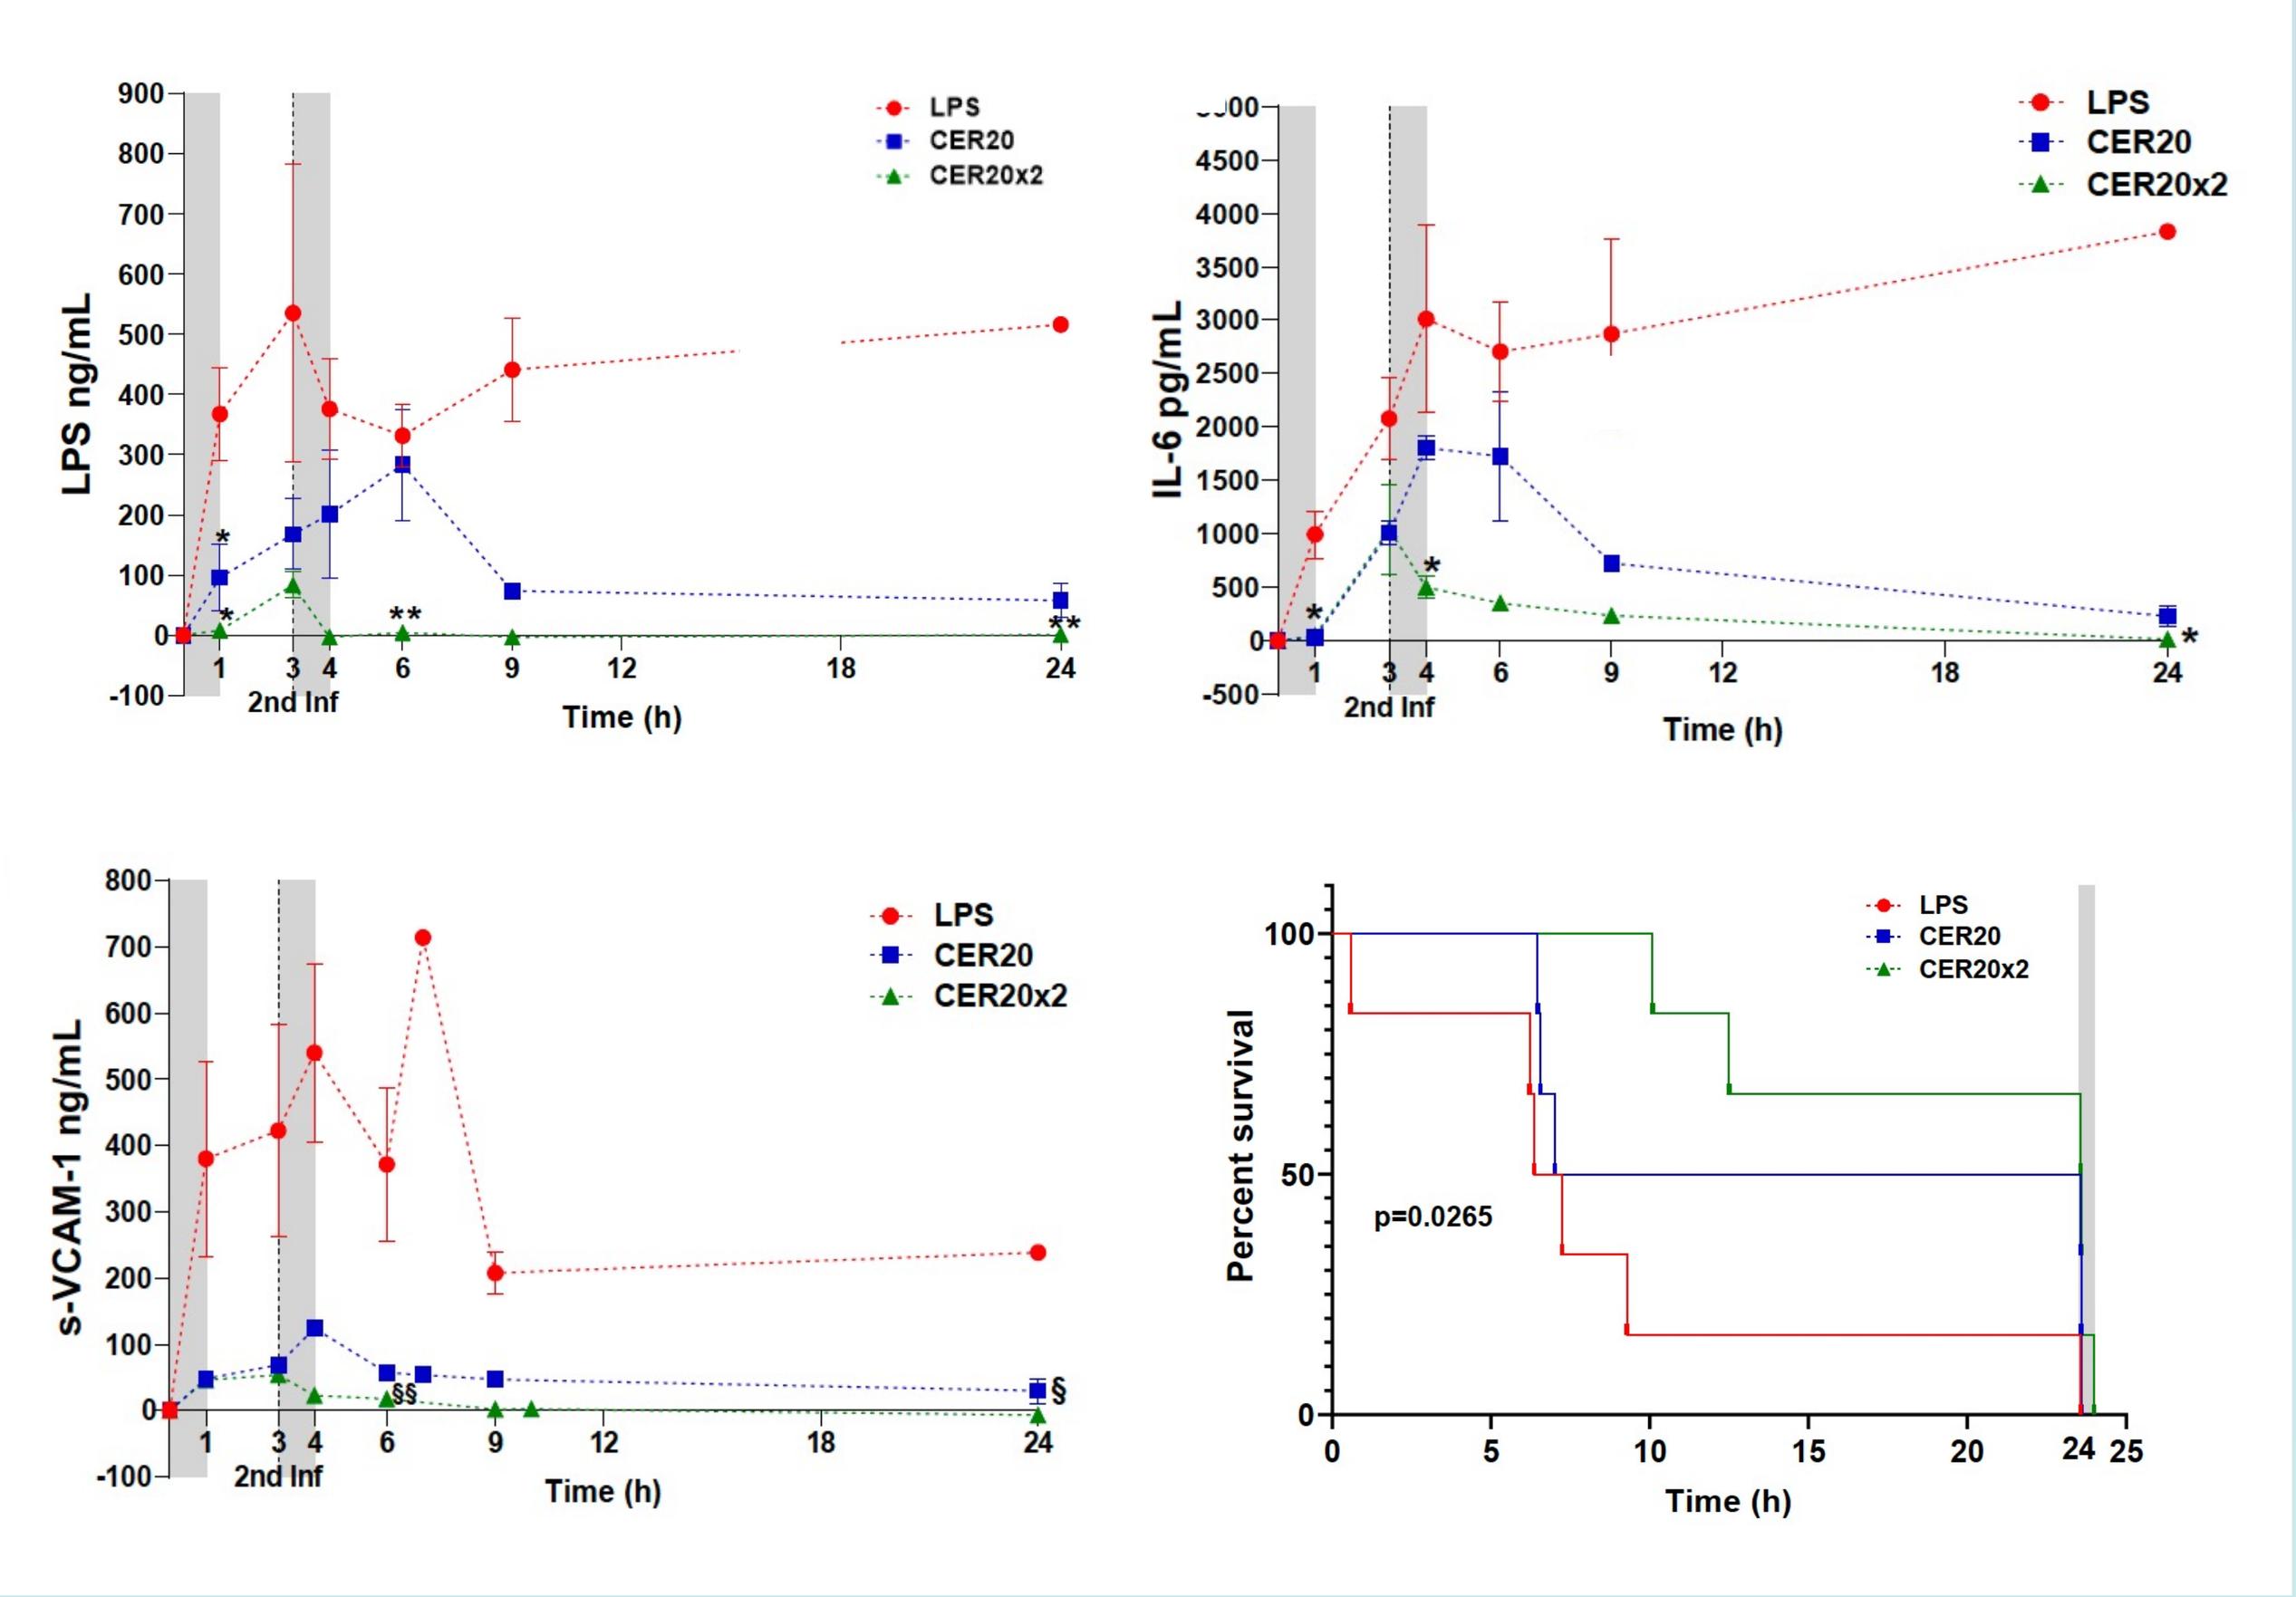
LPS ng/mL
IL-6 pg/mL
s-VCAM-1 ng/mL
Percent survival
Time (h)
2nd Inf
LPS
CER20
CER20x2
p=0.0265

Figure 1. *Decrease of systemic pro-inflammatory response and endothelial dysfunction and improved survival in a swine model of LPS-induced AKI.*

All animals received an infusion of LPS 300 ug/kg (T0) without (control group) or with infusion of CER-001 at 20 mg/kg (CER20 group) followed for half of this group by a second infusion at 3 h of 20 mg/kg of CER-001 (CER20x2 group).

Objective

We investigated the effects of CER-001 in a Phase 2a clinical trial, aiming to better understand its molecular basis in systemic inflammation and renal function.

Methods and Material

Open-label, randomized, dose-ranging (Phase 2a) study including 20 patients with sepsis due to intra-abdominal cavity infection or urosepsis, admitted at the Intensive Care Units (ICUs) and the Sub-intensive Nephrology Unit at A.O.U. Policlinico, Bari, Italy (N° EUDRACT 2020-004202-60, Protocol CER-001-SEP_AKI_01)(**Figure 2**).

Main Inclusion criteria

1) Sepsis sustained by Gram-negative bacteria receiving antibiotic treatment;
2) Met Sepsis 3 criteria;
3) Signed and dated informed consent provided by the patient or by a legal representative.

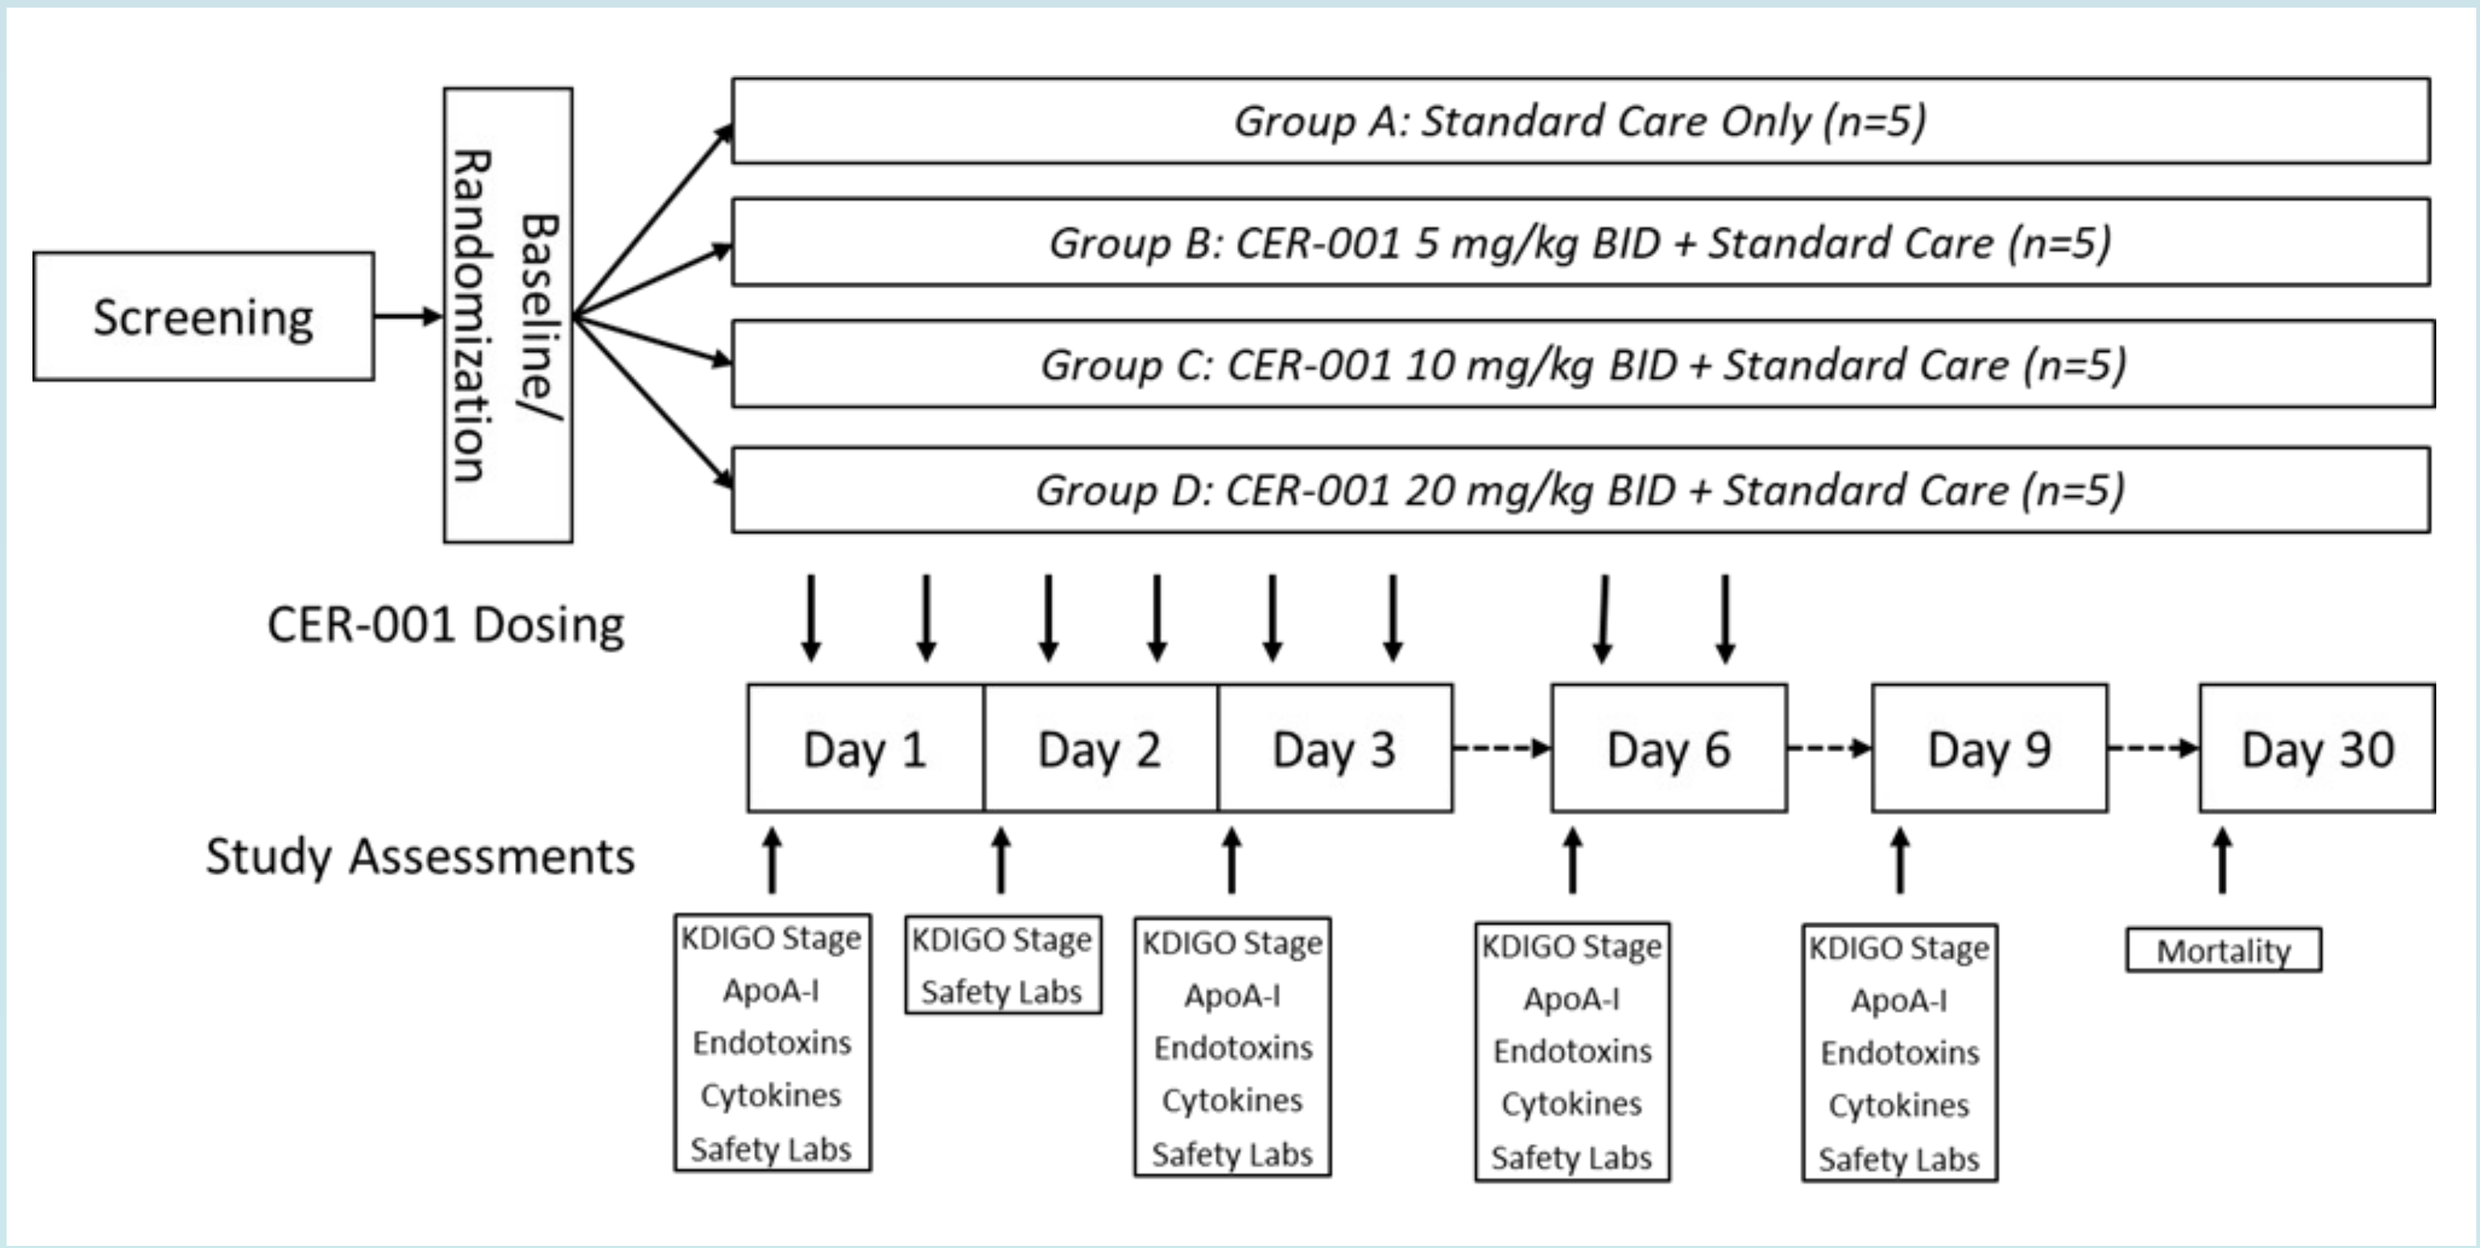
Screening
Baseline/ Randomization
Group A: Standard Care Only (n=5)
Group B: CER-001 5 mg/kg BID + Standard Care (n=5)
Group C: CER-001 10 mg/kg BID + Standard Care (n=5)
Group D: CER-001 20 mg/kg BID + Standard Care (n=5)
CER-001 Dosing
Day 1
Day 2
Day 3
Day 6
Day 9
Day 30
Study Assessments
KDIGO Stage ApoA-I Endotoxins Cytokines Safety Labs
KDIGO Stage Safety Labs
Mortality

Figure 2. *Treatment allocation and study interventions*

Study Objectives

- Determine an optimal dose of CER-001 in combination with standard of care in septic patients based on safety.
- Effects on AKI onset and severity
- Changes from baseline for endotoxin and IL-6 levels, and other key inflammatory and endothelial dysfunction markers.

Results

Rapid normalization of apoA-I levels with CER-001 was associated with significant and sustained LPS removal (p<0.05 on days 3, 6 and 9) and subsequent immunomodulation. CER-001 treatment led to rapid and significant decreases in pro-inflammatory cytokines (e.g., IL-6, IL-8, TNF-α, MCP-1), endothelial dysfunction markers (sVCAM, sICAM) and mortality biomarker (sTREM-1)(**Figure 3**).

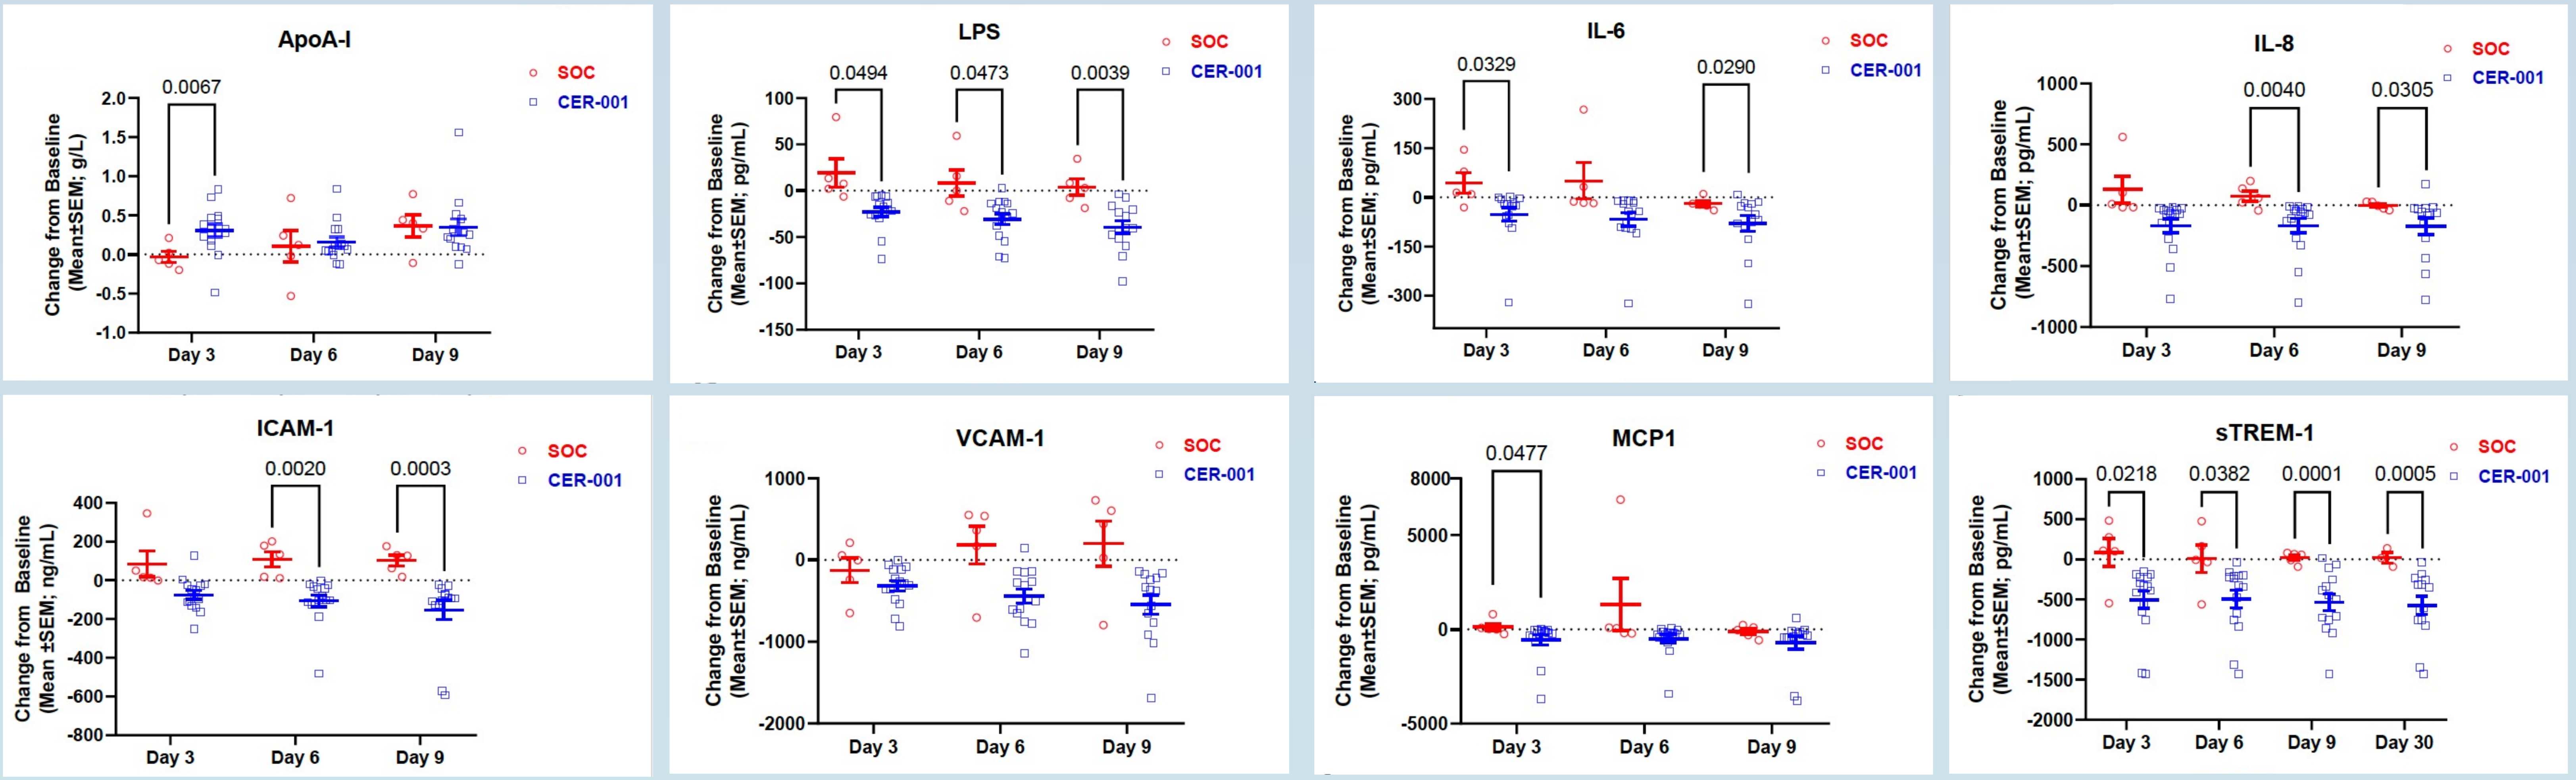
ApoA-I
LPS
IL-6
IL-8
ICAM-1
VCAM-1
MCP1
sTREM-1
SOC
CER-001
Change from Baseline (Mean±SEM)
Day 3
Day 6
Day 9
Day 30
0.0067
0.0494
0.0473
0.0039
0.0329
0.0290
0.0040
0.0305
0.0020
0.0003
0.0477
0.0218
0.0382
0.0001
0.0005

Figure 3*. Effect of CER-001 on ApoA-I level, LPS removal and inflammatory response in the pilot study RACERS*

CER-001-treated patients had a reduced risk of developing or progressing to severe AKI and a trend for improved survival. In a subset of critically ill patients, a shorter ICU stay with decreased need for organ support was observed with CER-001 (**Figure 4**).

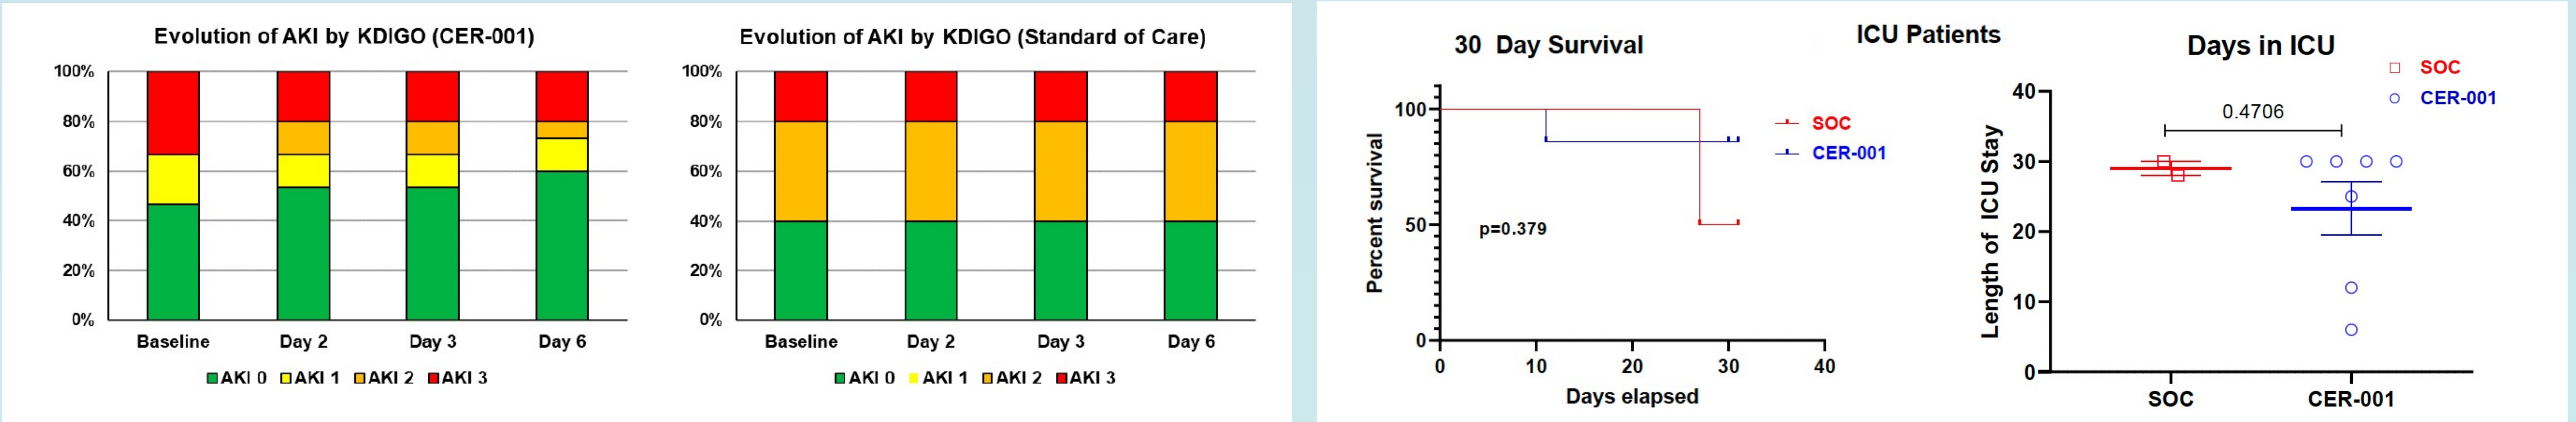
Evolution of AKI by KDIGO (CER-001)
Evolution of AKI by KDIGO (Standard of Care)
Baseline
Day 2
Day 3
Day 6
AKI 0
AKI 1
AKI 2
AKI 3
ICU Patients
30 Day Survival
Percent survival
Days elapsed
p=0.379
Days in ICU
Length of ICU Stay
0.4706
SOC
CER-001

Figure 4. *Effect of CER-001 on AKI risk and progression, length of stay and need for organ support in ICU patients*

Conclusions

CER-001 represents a promising therapeutic strategy for sepsis management, improving outcomes and mitigating the cytokine storm and associated organ damage often observed in our patients.

CONTACT INFORMATION: loreto.gesualdo@uniba.it

Publication will be available on Nov 2nd on BMC Medicine

BMC Medicine